TASECTAN™ (Gelatine Tannate) FOR THE TREATMENT OF ACUTE GASTROENTERITIS IN CHILDREN: PRELIMINARY RESULTS FROM A STUDY CONDUCTED TO EVALUATE ITS EFFICACY AND SAFETY.

Allegrini A[1], Costantini M[2]
[1]Oncology and Neurosciences Department - SS Annunziata Hospital of Chieti, Italia;
[2]Pharmacy Department - G. Mazzini Hospital of Teramo, Italia.

BACKGROUND

- Rotavirus is the leading cause for acute gastroenteritis (AGE) in infants and children associated with a substantial clinical and economic burden.
- Tasectan™ has been recently approved in Europe as a medical device to control and reduce the symptoms associated with diarrhoea. The use of gelatine tannate in children has been proved effective and safe in previous studies (Loeb H et al, 1989; Esteban Carretero et al, 2008).
- The study presented here provides additional evidence to support the use of Tasectan™ in children with AGE.

METHODS

- Open-label, non-randomised, non-controlled pilot study conducted in 19 children with AGE.
- **Inclusion criteria**: diagnosis of acute diarrhoea (≥3 watery stools 24 hours prior to first Tasectan™ dose) and a score of >20 mm using a 100-mm visual analogue pain scale.
- Exclusion criteria: ongoing treatment with other antidiarrhoeal at the time of inclusion in the study or known hypersensitivity to any of the active substances.
- Tasectan™ dosage: 1 sachet (250 mg) six times daily for two consecutive days.
- Clinical efficacy was evaluated in terms of reduction of both daily frequency of diarrhoeal stools and abdominal pain. Safety was assessed by recording AEs spontaneously reported by the patients.

RESULTS

Daily frequency of diarrhoeal stools

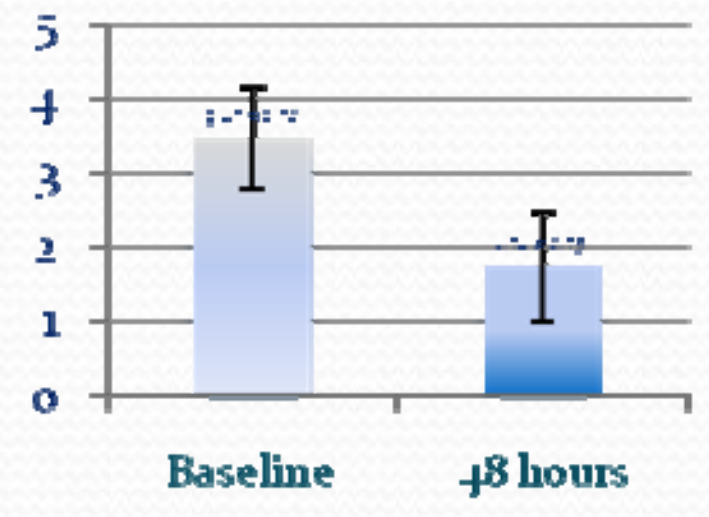
5
4
3
2
1
0
Baseline
48 hours

Abdominal pain

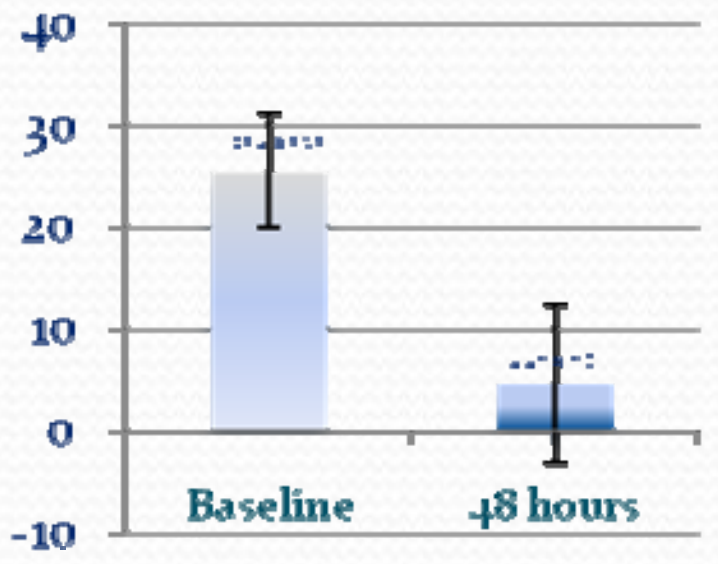
40
30
20
10
0
-10
Baseline
48 hours

The difference between the basal and final values for both efficacy outcomes was statistically significant. The treatment response rate was 89.5% at 48 hours (17 of 19 patients). No adverse effects were reported.

CONCLUSIONS

These preliminary results corroborate previous findings that gelatine tannate is effective and safe in children. This is particularly prescient as recent guidelines have discouraged the use of loperamide in children due to its serious adverse effects, which may outweigh the benefits particularly for under 3's.

Corresponding author: Alessandro Allegrini, Oncology and Neurosciences Department, SS Annunziata Hospital, 66100 – Chieti, Italia. E-mail: allegrinia@gmail.com